P294

Novel Design of a Focal Epilepsy Proof-of-Concept Study of RAP-219, a Negative Allosteric Modulator of the γ8 Transmembrane AMPA Receptor Associated Protein (TARPγ8)

William W Motley, MD[1]; Daniel Friedman, MD[2]; Kathryn A Davis, MD[3]; Bradley S Galer, MD[1]; Nancy Wyant, BA[1]; Jose A. Matta, PhD[4]; Arnold R Gammaitoni, PharmD[1]; Dennis Dlugos, MD, MSCE[5]; Martha Morrell, MD[6]; Jacqueline A French, MD[7]

[1]Rapport Therapeutics, Inc., Boston, MA, USA; [2]New York University Grossman School of Medicine, NYU Langone Health, New York, NY, USA; [3]University of Pennsylvania, Philadelphia, PA, USA; [4]Rapport Therapeutics, Inc., San Diego, CA, USA; [5]Children's Hospital of Philadelphia, University of Pennsylvania, Philadelphia, PA, USA; [6]Neuropace, Mountain View, CA, USA; [7]New York University Comprehensive Epilepsy Center, New York, NY, USA.

BACKGROUND

RAP-219: A novel mechanism of action

- α-Amino-3-hydroxy-5-methyl-4-isoxazolepropionic acid receptors (AMPARs) are a clinically validated therapeutic target for epilepsy
- Transmembrane AMPAR-associated regulatory proteins (TARPs) mediate AMPAR trafficking, subcellular localization, and gating[1]
- TARPγ8 has distinct regional expression in the central nervous system, enriched in the hippocampus, cortex, and amygdala with little to no expression in the hindbrain[1,2] (**Figure 1**)
- RAP-219 is a novel, potent, and selective negative allosteric modulator that binds at the interface between AMPAR and TARPγ8[3] (**Figure 2**)
- RAP-219 is well tolerated and efficacious in preclinical models of seizures (**Figure 3**)

Figure 1. Clinical PET Image of TARPγ8 Expression

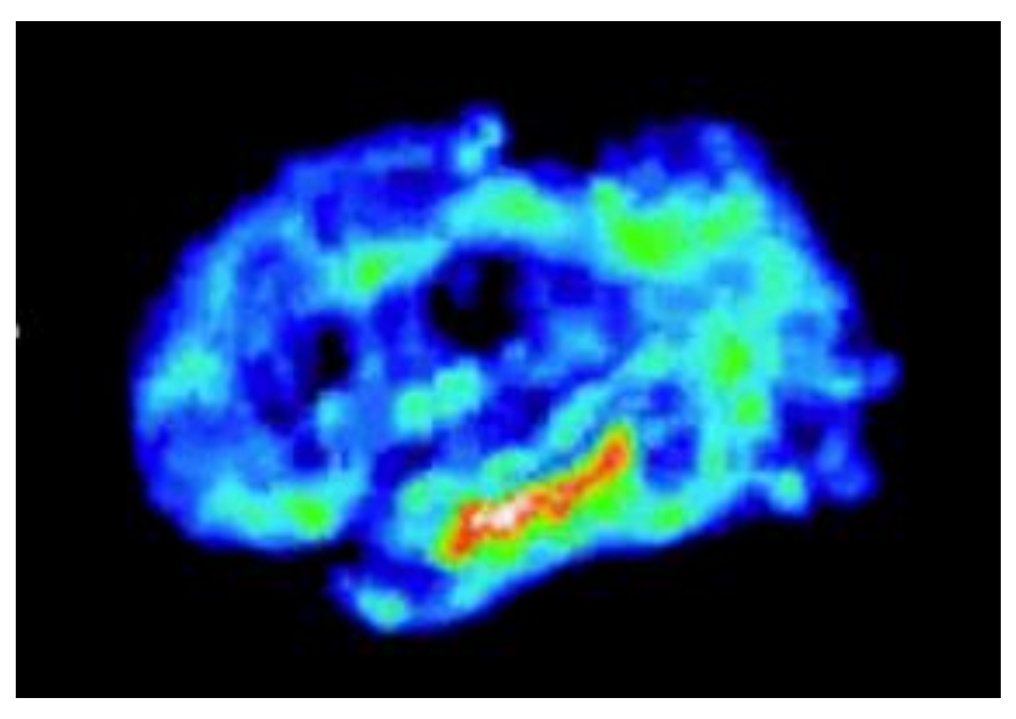

Radiotracer [[18]F]JNJ-64511070 targets AMPAR/TARPγ8.
AMPAR, AMPA receptor; PET, positron emission tomography; TARP, transmembrane AMPAR-associated regulatory protein.

Figure 2. TARPγ8 on AMPA Receptor Interface Mediates Drug Binding

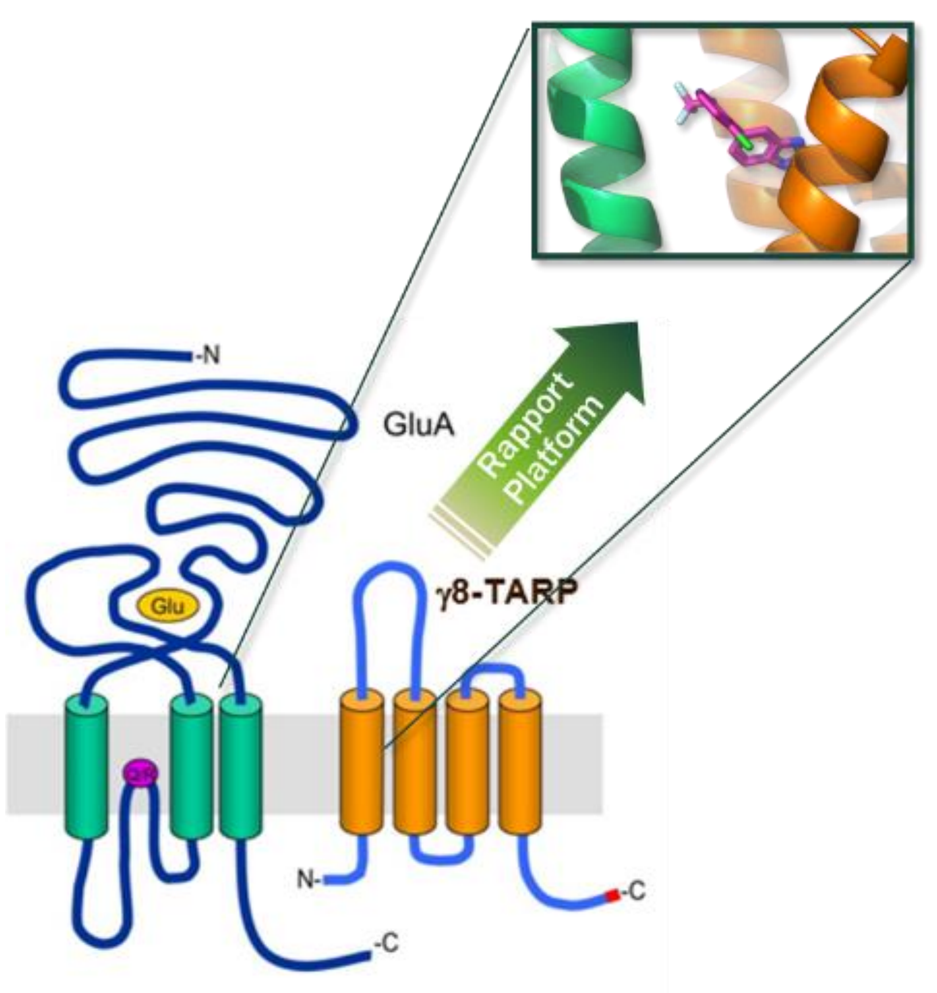
GluA
Rapport Platform
Glu
γ8-TARP
N
C

Figure 3. RAP-219 Improves Seizure Outcomes in Preclinical Models without Motoric Impairment

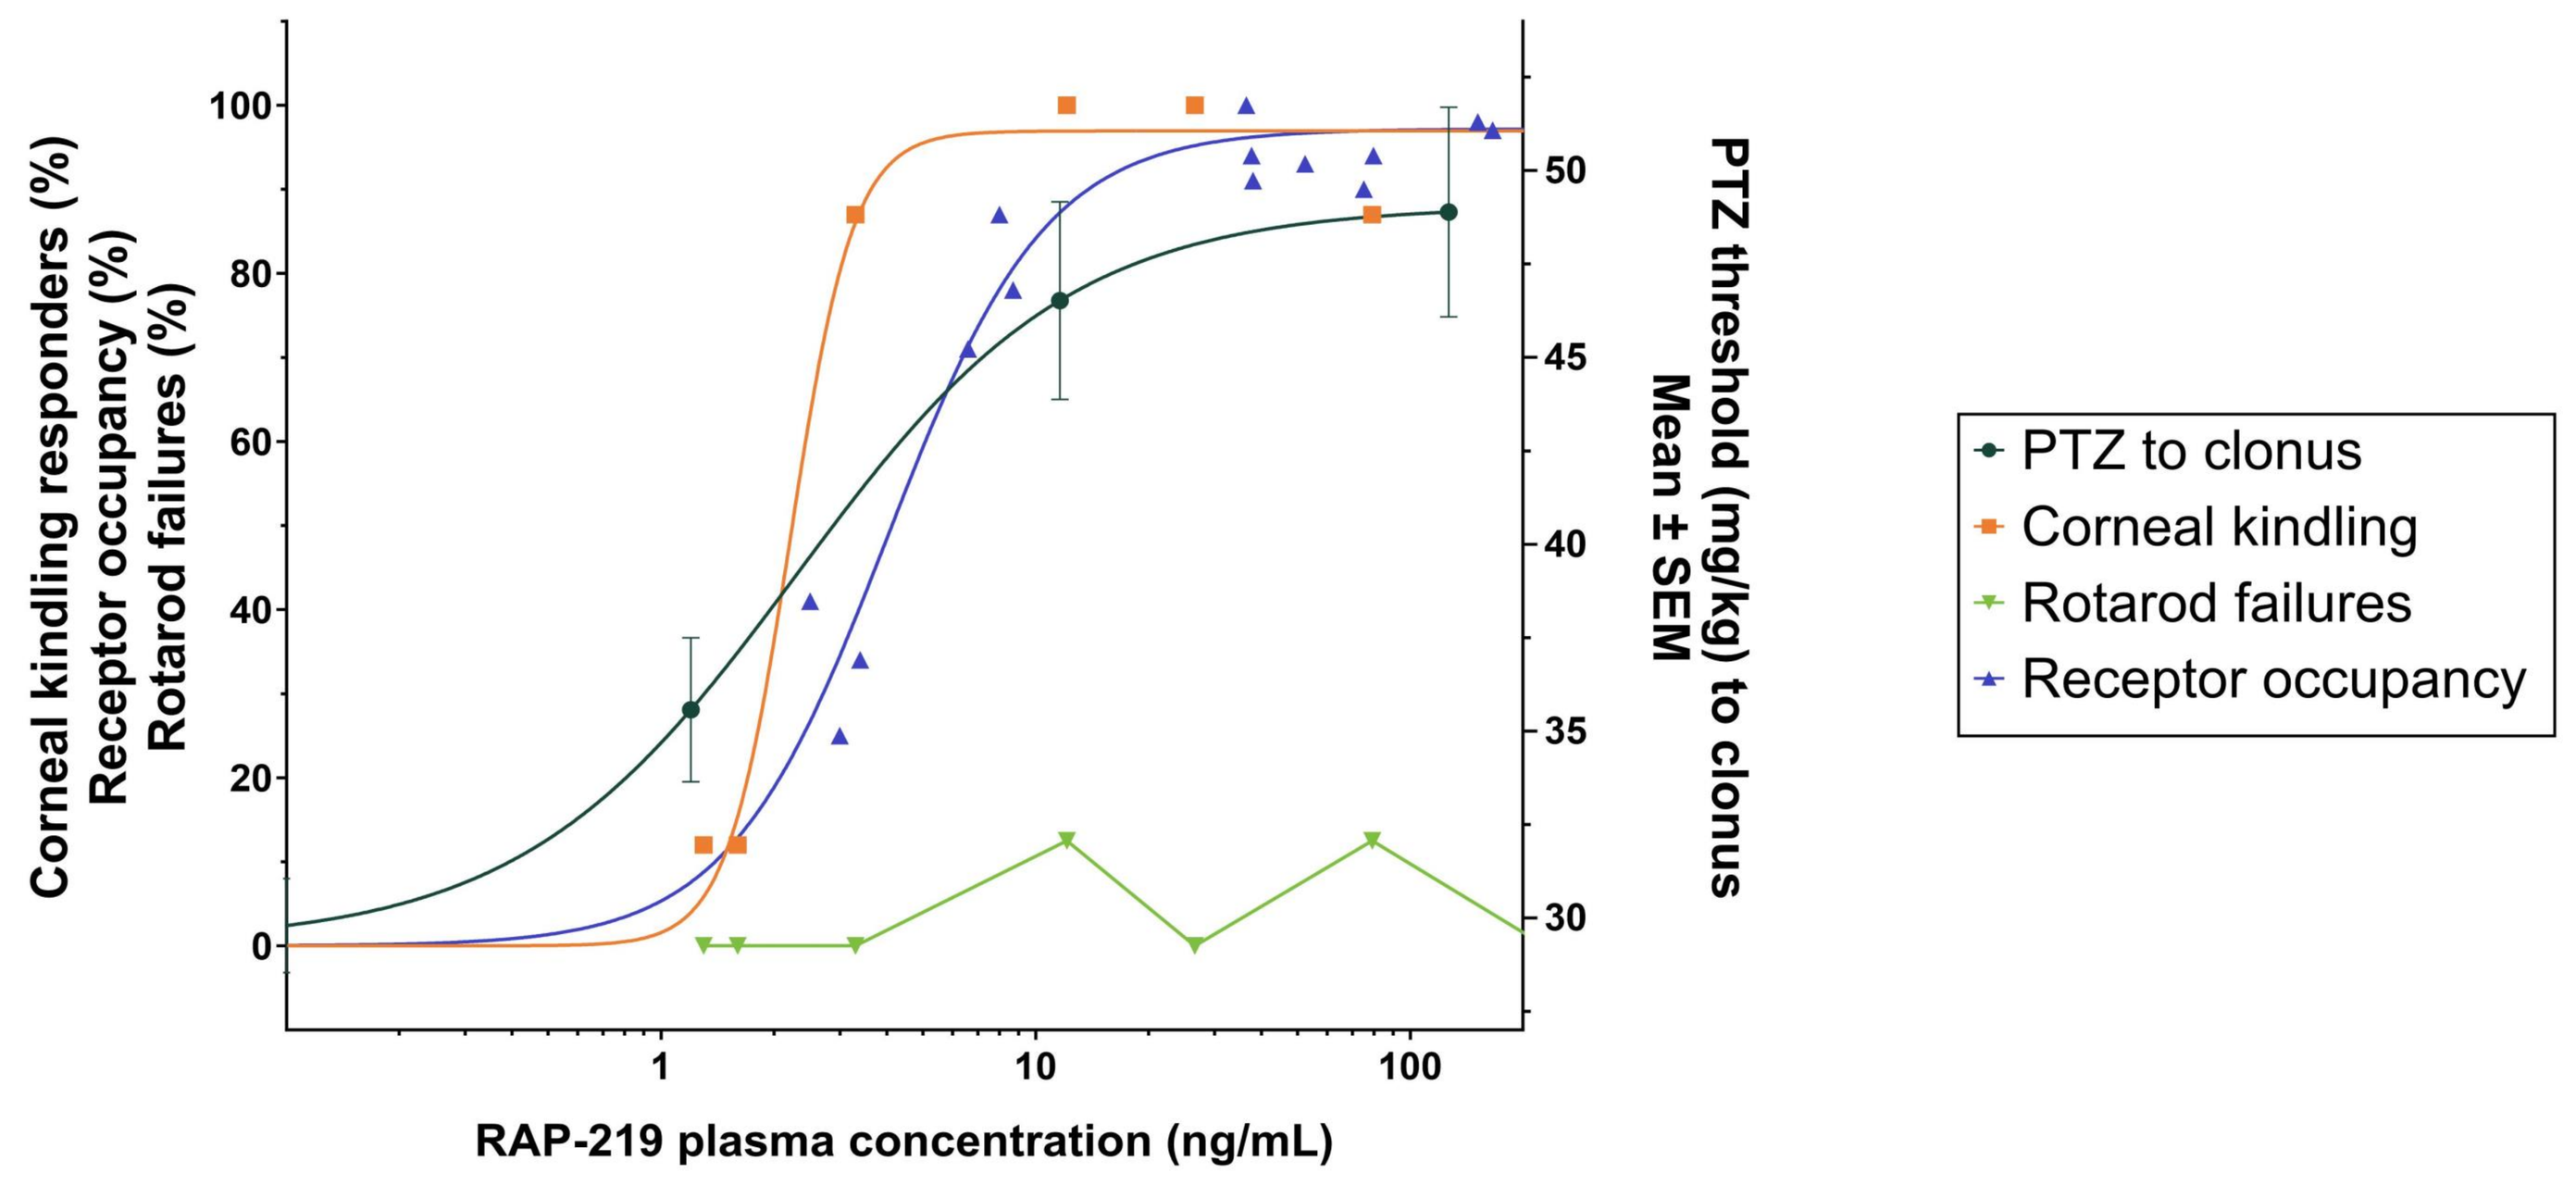
Corneal kindling responders (%)
Receptor occupancy (%)
Rotarod failures (%)
PTZ threshold (mg/kg) to clonus
Mean ± SEM
RAP-219 plasma concentration (ng/mL)
PTZ to clonus
Corneal kindling
Rotarod failures
Receptor occupancy
0
20
40
60
80
100
30
35
40
45
50
1
10
100

Receptor occupancy as a function of plasma concentration in rat at t=4h was used as a surrogate for this analysis, as potency in mouse vs rat was nearly identical (mouse pIC_{50}=9.8; rat pIC_{50}=9.9). PTZ threshold, corneal kindling, and rotarod were assessed in mouse.
PTZ, pentylenetetrazol.

Responsive neurostimulator: A tool for proof-of-concept study design

- RAP-219 is in phase 2A trial for the treatment of medically refractory focal onset seizures (FOS) using a novel proof-of-concept (POC) design
- A responsive neurostimulator (RNS® System, NeuroPace, Inc.) continually monitors electrographic activity from electrodes (intracranial encephalogram; iEEG) placed directly into the seizure focus or foci and is programmed by the patient's physician to detect epileptiform activity of significance including long episodes (LEs)
- LEs are:
 - Organized epileptiform activity exceeding a specified duration, typically 30 sec
 - Often represent electrographic seizures (EES)
 - A recognized seizure biomarker demonstrated to predict antiseizure medication response[4,5]
 - Utilized in this POC study of RAP-219 to provide efficacy signal detection and enable rapid progression into registrational trials (**Table 1**)

Table 1. POC Study Designs for Drug-Resistant FOS

Model Attributes	Responsive neurostimulator (RNS® System)	Photosensitivity	Transcranial magnetic stimulation (TMS)
Uses focal epilepsy patient population	Yes	No	No
Direct measure of drug activity	Yes Reduction in LEs	No Evoked generalized epileptiform discharges	No Provoked cortical hyperexcitability
Informs dose selection for registrational trials	Yes PK/PD data can provide measure of efficacy degree at different exposure levels	No Indirect dose response readout	No Indirect dose response readout
Enables rapid progression into registrational trials	Yes Biomarker data to inform dose and effect size	No Does not inform dose or effect size	No Does not inform dose or effect size

FOS, focal onset seizures; LE, long episode; PK/PD, pharmacokinetic/pharmacodynamic; POC, proof-of-concept.

- iEEG measures (i.e., LEs, detection counts, spike rate) have been shown to correlate with clinically meaningful seizure frequency reduction (≥50% reduction)[4,5]
 - LE frequency reduction demonstrated the strongest correlation of the iEEG measures[5]
- A 30-40% reduction in LEs within 1-4 weeks of new ASM initiation was associated with >50% seizure reduction[6]

METHODS

Figure 4. Study Design

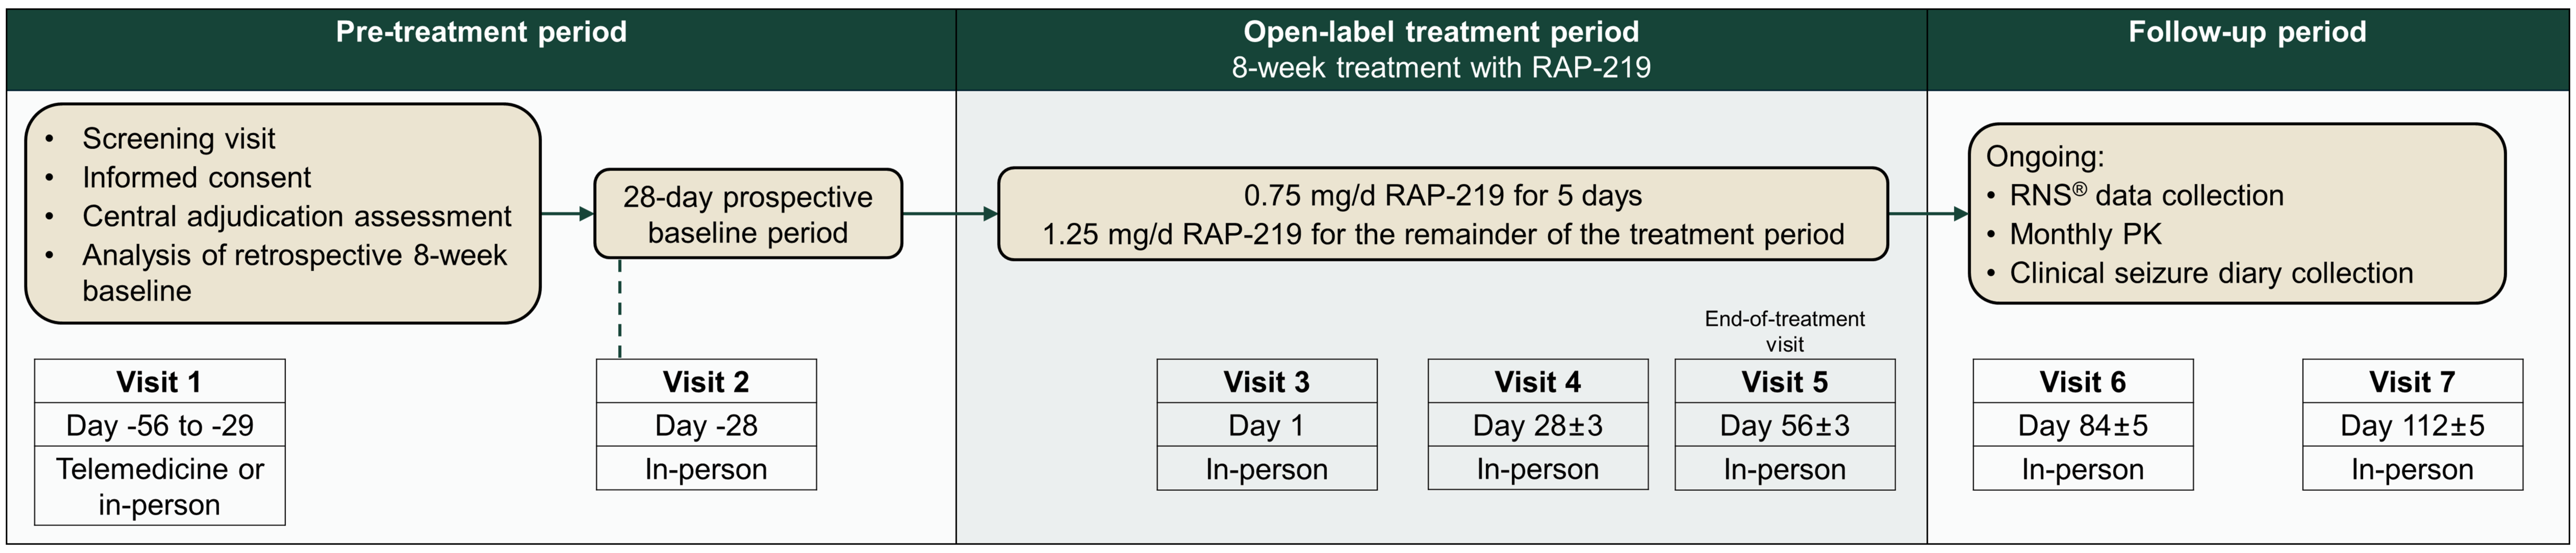
Pre-treatment period
Open-label treatment period
8-week treatment with RAP-219
Follow-up period
Screening visit
Informed consent
Central adjudication assessment
Analysis of retrospective 8-week baseline
28-day prospective baseline period
0.75 mg/d RAP-219 for 5 days
1.25 mg/d RAP-219 for the remainder of the treatment period
Ongoing:
RNS® data collection
Monthly PK
Clinical seizure diary collection
End-of-treatment visit
Visit 1
Day -56 to -29
Telemedicine or in-person
Visit 2
Day -28
In-person
Visit 3
Day 1
In-person
Visit 4
Day 28±3
In-person
Visit 5
Day 56±3
In-person
Visit 6
Day 84±5
In-person
Visit 7
Day 112±5
In-person

Approximately 20 patients with an implanted RNS® device are expected to receive RAP-219 during this multicenter, open-label POC study.
PK, pharmacokinetic; POC, proof-of-concept; RNS®, responsive neurostimulator.

Approximately 20 patients meeting the following key inclusion criteria:
18-65 years old
Medically refractory FOS
An implanted RNS® device and: • RNS® implanted ≥15 months before screening • Stable device configuration, stimulation, and detection settings, including LE duration • An average of ≥8 LEs per 4-week interval during the combined retrospective/prospective baseline periods • Concordance of ≥50% between LEs and EES • No anticipated need for device setting changes during the study period
≥1 clinical seizure(s) during the retrospective baseline period
Key exclusion criteria:
Use of perampanel within 12 weeks before screening

EES, electrographic seizures; FOS, focal onset seizures; LE, long episode; RNS®, responsive neurostimulator.

Key endpoints
LE frequency responder analysis (% of patients that demonstrate ≥30% reduction in LEs)
Change in LE frequency per 28 days during the second 4-week interval of open-label treatment period compared to frequency across retrospective and prospective baselines
Change in estimated EES, clinical seizure frequency, and additional iEEG biomarkers per 28 days during the second 4-week interval of open-label treatment period compared to frequency across retrospective/ prospective baseline periods
Number of patients with clinically meaningful improvement in PGIC and/or CGIC measures
Incidence of TEAEs and SAEs

CGIC, Caregiver Global Impression of Change; EES, electrographic seizures; iEEG, intracranial electroencephalogram; LE, long episode; PGIC, Patient Global Impression of Change; SAEs, serious adverse events; TEAEs, treatment-emergent adverse events.

CONCLUSIONS

- This novel POC study will enable evaluation of RNS® System measures to provide efficacy signals (biomarkers) to inform later phases of clinical development
- Analyses included in the POC study will offer insight into the efficacy of RAP-219, a novel treatment targeting TARPγ8 as a precision approach for management of FOS

REFERENCES

1. Coombs ID, et al. *Mol Pharmacol.* 2022;101(5):343-56.
2. Maher MP, et al. *J Pharmacol Exp Ther.* 2016;357(2):394-414.
3. Yu J, et al. Nature. 2021;594(7863):445-453.
4. Quraishi IH, et al. *Epilepsia.* 2020;61(1):138-48.
5. Skarpaas TL, et al. *Epilepsy Behav.* 2018;83:192-200.
6. Rapport, Data on file.

ACKNOWLEDGMENTS

The authors thank Mari Willeman, PhD, and Anthony DiLauro, PhD, of PharmaWrite, LLC, for medical writing and editorial assistance, which were funded by Rapport Therapeutics, Inc.

DISCLOSURES

WWM, BSG, NW, JAM, ARG: Rapport Therapeutics: employee, stock ownership; **DF**, Epilepsy Study Consortium: research services from Axcella, Biogen, Cerebel, Crossject, Engage Pharmaceuticals, Eisai, Pfizer, SK Life Science, Xenon, and Zynerba. Consultant: Eisai, Neurelis Pharmaceuritcals. Travel support: Medtronics, Eisai, Epilepsy Foundation. Research support: CDC, NINDS, Epilepsy Foundation, Empatica, Epitel, UCB Inc, Neuropace. Scientific advisory board: Receptor Life Sciences. Equity interests: Neuroview Technology, Receptor Life Sciences. **KAD:** Rapport Therapeutics: consultant. **DD:** Epilepsy Study Consortium: consulting, advisory boards/investigator meetings for Beacon Biosignals, Biohaven Pharmaceuticals, Encoded Therapeutics, Grin Therapeutics, Jazz Pharmaceuticals, Longboard Pharmaceuticals, Rapport Therapeutics, SK Life Sciences, Stoke, Takeda, and UCB. **JAF:** Epilepsy Foundation: consultant; Epilepsy Study Consortium: President, consulting, advisor boards.

Please follow the QR code for a copy of the poster

Presented at the 15th European Epilepsy Congress, September 7-11, 2024, Rome, Italy